RESILIENT: study of irinotecan liposome injection (nal-IRI) in patients with small cell lung cancer: preliminary findings from part 1 dose-defining phase

Luis G Paz-Ares,[1] David R Spigel,[2] Christoph Zielinski,[3] Yuanbin Chen,[4] Maria Jove,[5] Oscar Juan-Vidal,[6] David Chu,[7] Patricia Rich,[8] Theresa Hayes,[9] Maria Gutierrez Calderon,[10] Reyes Bernabe Caro,[11] Alejandro Navarro Mendivil,[12] Afshin Dowlati,[13] Bin Zhang,[14] Yan Moore,[14] Tiffany Wang,[14] Natalya Nazarenko,[14] Paul Bunn[15]

[1]Hospital Universitario 12 de Octubre, Madrid, Spain; [2]Sarah Cannon Research Institute, Tennessee Oncology, Nashville, TN, USA; [3]Vienna Cancer Center and Central European Cooperative Oncology Group, Vienna, Austria; [4]Cancer & Hematology Centers of Western Michigan, Grand Rapids, MI, USA; [5]Institut Català d'Oncologia, Hospital Duran i Reynals, Barcelona, Spain; [6]Hospital Universitari i Politècnic La Fe, Valencia, Spain; [7]North Shore Hematology Oncology Associates, PC, Bay Shore, NY, USA; [8]Cancer Treatment Centers of America, Atlanta, GA, USA; [9]South West Healthcare, VIC, Australia; [10]Hospital Regional Universitario de Málaga, Malaga, Spain; [11]Hospital Universitario Virgen del Rocio, Seville, Spain; [12]Vall d'Hebron Barcelona Hospital, Barcelona, Spain; [13]Case Western Reserve University, Cleveland, OH, USA; [14]Ipsen Bioscience, Boston, MA, USA; [15]Cancer Center and Department of Medicine, University of Colorado, Denver, CO, USA

TBC

BACKGROUND

- In the treatment of small cell lung cancer (SCLC), the mainstay first-line (1L) treatment is combination chemotherapy, typically involving platinum-based therapy (carboplatin or cisplatin) plus etoposide.[1,2]
 - Although SCLC is often sensitive to chemotherapy and radiotherapy, many patients relapse and develop drug resistance.[2,3]
- Irinotecan is an inhibitor of topoisomerase 1, a validated drug target in SCLC.
- Liposomal irinotecan (nal-IRI) is a liposomal formulation of irinotecan, designed to use the enhanced permeability and retention effect to achieve liposomal deposition in tumor tissue followed by local irinotecan release and subsequent conversion to the active metabolite SN-38.
- nal-IRI is approved for the treatment of metastatic adenocarcinoma of the pancreas after progression following a gemcitabine-based therapy.[4]
- RESILIENT is an ongoing phase II/III trial assessing the safety, tolerability and efficacy of nal-IRI injection in patients with SCLC, comprising part 1 (dose-finding and -expansion phase) and part 2 (randomized controlled trial).
- Here, we report data from the dose-finding phase of part 1 of the RESILIENT trial.

METHODS

Study design

- RESILIENT (NCT03088813) is a two-part phase II/III study (**Figure 1**).
- Part 1 of the study is an open-label, single-arm, dose-finding and -expansion phase with the aim of:
 - determining the recommended nal-IRI monotherapy dose for part 2 of the study
 - defining the safety and tolerability of nal-IRI and demonstrating the preliminary efficacy of nal-IRI monotherapy administered every 2 weeks.
- Part 2 is a randomized, controlled, efficacy-assessment phase with the aim of comparing efficacy and safety profiles of nal-IRI monotherapy and topotecan monotherapy.

Figure 1. RESILIENT study design

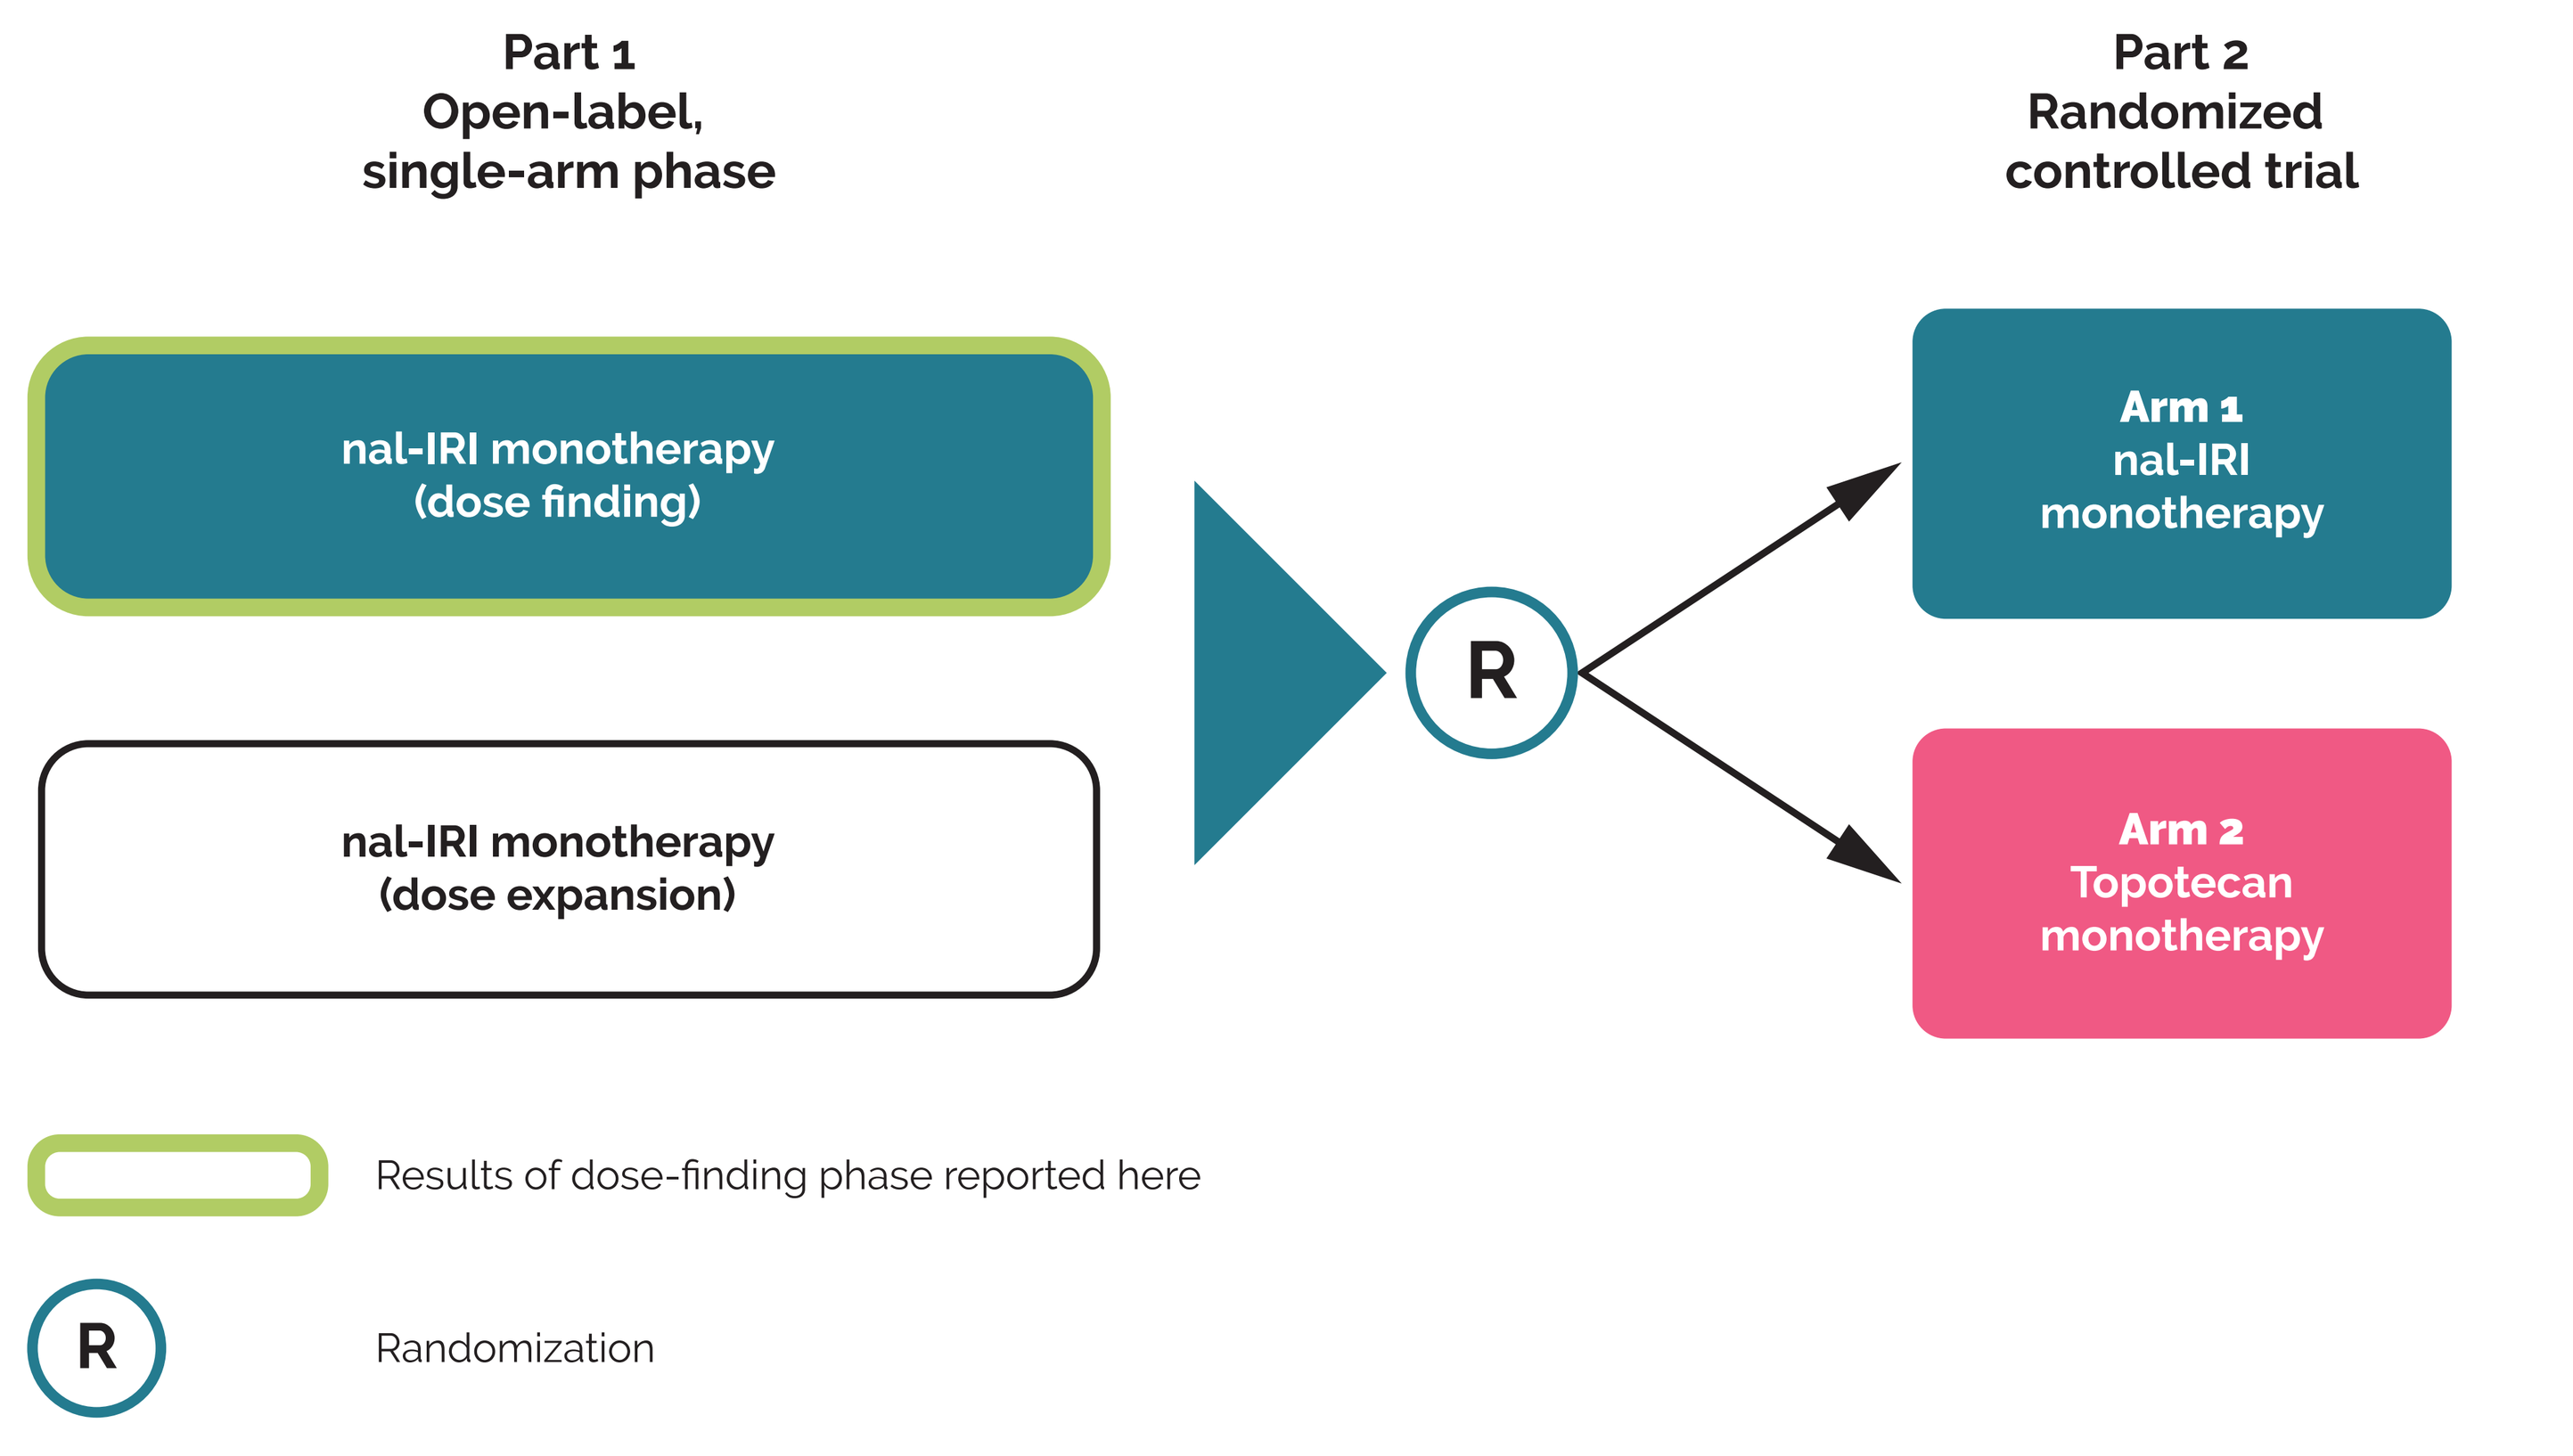
Part 1
Open-label,
single-arm phase
Part 2
Randomized
controlled trial
nal-IRI monotherapy
(dose finding)
nal-IRI monotherapy
(dose expansion)
R
Arm 1
nal-IRI
monotherapy
Arm 2
Topotecan
monotherapy
Results of dose-finding phase reported here
R
Randomization

nal-IRI, liposomal irinotecan

Patients

- Adults (≥ 18 years old) with SCLC who progressed on or after 1L platinum-based chemotherapy (carboplatin or cisplatin), immunotherapy or chemoradiation, including platinum-based chemotherapy for treatment of limited or extensive-stage SCLC.
- Additional inclusion criteria
 - Eastern Cooperative Oncology Group performance status of 0 or 1 and adequate organ function.
 - Prior exposure to immunotherapy is allowed.
 - Asymptomatic central nervous system metastases are allowed.

Treatment during the dose-finding phase of RESILIENT part 1

- nal-IRI at a dose of 85 mg/m² is administered intravenously over 90 minutes every 2 weeks (cohort 1 [C-1]).
- If nal-IRI 85 mg/m² is deemed to have unacceptable toxicity, a dose of 70 mg/m² administered over 90 minutes every 2 weeks is recommended as a low-dose cohort contingency (cohort 2 [C-2]).

Endpoints of the dose-finding phase of RESILIENT part 1

- Primary endpoints are safety and tolerability at the 85 mg/m² and 70 mg/m² dose levels.
- Secondary endpoints are objective response rate (ORR), progression-free survival (PFS) and overall survival (OS).
- For efficacy endpoints, tumor assessment (by the investigator) is performed at screening and every 6 weeks (± 1 week) from cycle 1, day 1, and in accordance with Response Evaluation Criteria in Solid Tumors (RECIST) v1.1.

RESULTS OF THE DOSE-FINDING PHASE OF RESILIENT PART 1

Patient disposition and baseline characteristics

- As of December 24, 2018, 12 patients have received at least one dose of nal-IRI: four patients in C-1 (85 mg/m²) and eight patients in C-2 (70 mg/m²).
- Baseline characteristics are summarized in **Table 1**.

Table 1. Baseline characteristics

Characteristic[a]	nal-IRI Cohort 1 85 mg/m² (n = 4)	nal-IRI Cohort 2 70 mg/m² (n = 8)	nal-IRI Total (N = 12)
Sex, n (%)			
Male	2 (50.0)	3 (37.5)	5 (41.7)
Female	2 (50.0)	5 (62.5)	7 (58.3)
Age, years			
Median	61.5	58.0	60.0
Range	59–63	49–73	49–73
Race, n (%)			
White	4 (100.0)	8 (100.0)	12 (100.0)
ECOG performance status, n (%)			
0	1 (25.0)	1 (12.5)	2 (16.7)
1	3 (75.0)	7 (87.5)	10 (83.3)
Previous chemotherapy cycles, n (%)			
4	0	1 (12.5)	1 (8.3)
≥ 6	4 (100.0)	7 (87.5)	11 (91.7)
Best response, n (%)			
CR	0	1 (12.5)	1 (8.3)
PR	2 (50.0)	4 (50.0)	6 (50.0)
SD	1 (25.0)	1 (12.5)	2 (16.7)
PD	1 (25.0)	2 (25.0)	3 (25.0)

[a]Only categories for which non-zero values were recorded are shown. CR, complete response; ECOG, Eastern Cooperative Oncology Group; nal-IRI, liposomal irinotecan; PD, progressive disease; PR, partial response; SD, stable disease

- All four patients in C-1 discontinued study treatment (two owing to disease progression, one owing to a study drug-related adverse event [AE] and one owing to death).
- Two patients in C-2 discontinued study treatment (both owing to disease progression).
- One patient in each cohort died before completion of the dose-finding phase of the study.
- As of December 24, 2018, eight patients were continuing to participate in the study (one in C-1 and seven in C-2).

Safety and tolerability

- Mean (standard deviation) of the total actual dose received was 459.8 mg (278.7 mg) and 714.8 mg (331.3 mg) in C-1 and C-2, respectively.
- The most commonly reported treatment-emergent AEs of any grade were diarrhea (91.7%), nausea (66.7%), decreased appetite (58.3%), fatigue (41.7%) and vomiting (41.7%) (**Table 2**).
- Serious AEs occurred in 3 of 4 patients in C-1 and 1 of 8 patients in C-2.
 - Serious AEs considered to be related to study treatment occurred in two patients (both in C-1; one report each of diarrhea, nausea, vomiting, hepatotoxicity and abnormal liver function test).
- Four patients in C-1 had dose-limiting toxicities (diarrhea, n = 3; abnormal liver function test, n = 1; hyponatremia, n = 1); there were no dose-limiting toxicities in C-2.

Table 2. Treatment-emergent adverse events occurring in > 1 patient in total

Preferred term	nal-IRI Cohort 1 85 mg/m² (n = 4)	nal-IRI Cohort 2 70 mg/m² (n = 8)	nal-IRI Total (N = 12)
Diarrhea	4 (100.0)	7 (87.5)	11 (91.7)
Nausea	3 (75.0)	5 (62.5)	8 (66.7)
Decreased appetite	4 (100.0)	3 (37.5)	7 (58.3)
Fatigue	2 (50.0)	3 (37.5)	5 (41.7)
Vomiting	1 (25.0)	4 (50.0)	5 (41.7)
Abdominal pain	1 (25.0)	3 (37.5)	4 (33.3)
Hypokalemia	1 (25.0)	3 (37.5)	4 (33.3)
Asthenia	1 (25.0)	2 (25.0)	3 (25.0)
Constipation	2 (50.0)	1 (12.5)	3 (25.0)
Hypomagnesemia	1 (25.0)	2 (25.0)	3 (25.0)
Anemia	2 (50.0)	0	2 (16.7)
Arthralgia	2 (50.0)	0	2 (16.7)
Dehydration	0	2 (25.0)	2 (16.7)
Edema peripheral	1 (25.0)	1 (12.5)	2 (16.7)
Headache	1 (25.0)	1 (12.5)	2 (16.7)
Hypocalcemia	1 (25.0)	1 (12.5)	2 (16.7)
Neutropenia	2 (50.0)	0	2 (16.7)
Weight decreased	1 (25.0)	1 (12.5)	2 (16.7)

Data are number (%) of patients. Treatment-emergent AEs are AEs that occurred or worsened on or after the day of the first dose of study medication and in the 30 days after discontinuation of the study drug. Patients with multiple AEs within a system organ class or preferred term were only counted once under each category.
AE, adverse event; nal-IRI, liposomal irinotecan

Efficacy

- The ORR for the total study population was 33.3% (**Table 3**).
 - A higher ORR was achieved with the 70 mg/m² dose than with the 85 mg/m² dose (37.5% vs 25.0%, respectively).

Table 3. Objective response rates

Category	nal-IRI Cohort 1 85 mg/m² (n = 4)	nal-IRI Cohort 2 70 mg/m² (n = 8)	nal-IRI Total (N = 12)
Best objective response, n (%)			
CR	0	0	0
PR	1 (25.0)	3 (37.5)	4 (33.3)
SD	1 (25.0)	2 (25.0)	3 (25.0)
PD	1 (25.0)	1 (12.5)	2 (16.7)
Non-evaluable	1 (25.0)	2 (25.0)	3 (25.0)
ORR, n (%)			
CR+PR	1 (25.0)	3 (37.5)	4 (33.3)
Non-responder (censored)	3 (75.0)	5 (62.5)	8 (66.7)

CR, complete response; NE, not estimable; OR, objective response; ORR, objective response rate; PD, progressive disease; PR, partial response; SD, stable disease

CONCLUSION

- Initial assessment suggests that nal-IRI at a dose of 70 mg/m² administered on a 2-weekly basis is well tolerated and has promising antitumor activity in patients with SCLC who progressed on or after a platinum-based regimen.
- The part 1 dose-expansion phase is ongoing.

References

1. Morabito A *et al. Crit Rev Oncol Hematol* 2014;9:257–70
2. Früh M *et al. Ann Oncol* 2013;24(Suppl 6):vi99–105
3. Simon GR *et al. Chest* 2007;132(Suppl 3):324S–39S
4. ONIVYDE (irinotecan liposome injection) for intravenous use – prescribing information. 2015. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2015/207793lbl.pdf (Accessed May 2019)

Disclosures

LGPA – holder honoraria: AstraZeneca, Boehringer Ingelheim, Bristol-Myers Squibb, Lilly, MSD and Roche.
DRS – consultant: AstraZeneca, Boehringer Ingelheim, Bristol-Myers Squibb, Celgene, Clovis Oncology, Genentech/Roche, Lilly, Novartis and Pfizer; grants/research support: Amgen, AstraZeneca, Boehringer Ingelheim, Bristol-Myers Squibb, Celgene, Clovis Oncology, Daiichi Sankyo, Genentech/Roche, Lilly, Merck, Novartis, OncoGenex, OncoMed Pharmaceuticals, Peregrine Pharmaceuticals, Pfizer, University of Texas Southwestern Medical Center – Simmons Comprehensive Cancer Center and Verastem Oncology; other: Bristol-Myers Squibb.
CZ – speaker/advisory board: ARIAD Pharmaceuticals, AstraZeneca, Boehringer Ingelheim, Bristol-Myers Squibb, FibroGen, Imugene, Merck KGaA, MSD, Novartis and Roche.
YC: TBC
MJ: TBC
OJV: TBC
DC: TBC
PR: TBC
TH: TBC
MGC: TBC
RBC: TBC
ANM: TBC
AD: TBC
BZ, YM, TW and NN – employment: Ipsen.
PB – consultant: AstraZeneca, Bristol-Myers Squibb, Daiichi Sankyo, Genentech, Lilly, Merck, Merrimack Pharmaceuticals, Novartis and Pfizer; other: AstraZeneca, Bristol-Myers Squibb, Genentech, Merck and Pfizer.

Acknowledgments

The authors thank all patients involved in the study, as well as their caregivers, care team, investigators and research staff in participating institutions.
The study was supported by Ipsen Biopharmaceuticals.
The authors thank Oxford PharmaGenesis, Oxford, UK, for providing medical writing support, which was sponsored by Ipsen Biopharmaceuticals, Inc., Basking Ridge, NJ, USA, in accordance with Good Publication Practice guidelines.
Contact: natalya.nazarenko@ipsen.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the authors of this poster.

QR code

Presented at ASCO 2019 | Chicago, IL, USA | May 31–June 4, 2019

This study was sponsored by Ipsen